PEP07: A novel, brain penetrant oral Chk1 inhibitor for the treatment of AML and MCL

Bettice Chen[1], Kyla Grimshaw[2], Jack Cheng[1], Allen Lee[1], Mel Liu[1], Meriel Major[2], Bob Boyle[2], Hong-Ren Wang[1]

Abstract

Chk1 is a key modulator of the cell division cycle and DNA damage response (DDR) signaling. Inhibition of Chk1, in conjunction with additional genetic alterations and/or addition of a DNA damaging agent, results in mitotic catastrophe and apoptosis of cancer cells, offering an attractive approach to treat cancer[3].

Acute myeloid leukemia (AML) is characterized by a deranged DDR pathway and high Chk1 expression that is associated with poor patient outcomes[4]. Mantle cell lymphoma (MCL) is a rare and aggressive form of Non-Hodgkin lymphoma with the genetic hallmark of a chromosomal translocation leading to the over expression of cyclin D1. Since Chk1 regulates cdk1/cyclin activity, Chk1 inhibitors have been proposed as a novel therapeutic approach in this cancer[5].

Here, we present PEP07, an orally available brain penetrant selective Chk1 inhibitor that is entering first in human clinical studies in AML and MCL.

In vitro properties of PEP07

PEP07 is a potent and selective Chk1 inhibitor with over 1000-fold selectivity vs Chk2. PEP07 treatment causes inhibition of Chk1 autophosphorylation and activation of the DDR and apoptotic pathways leading to cancer cell death.

a)

Target	IC50, nM
Chk1	1
Chk2	1630
Flt-3	37
Rsk3	42
Rsk2	83
RET	81
Rsk4	86
Rsk1	125
MAP4K4	242

b)

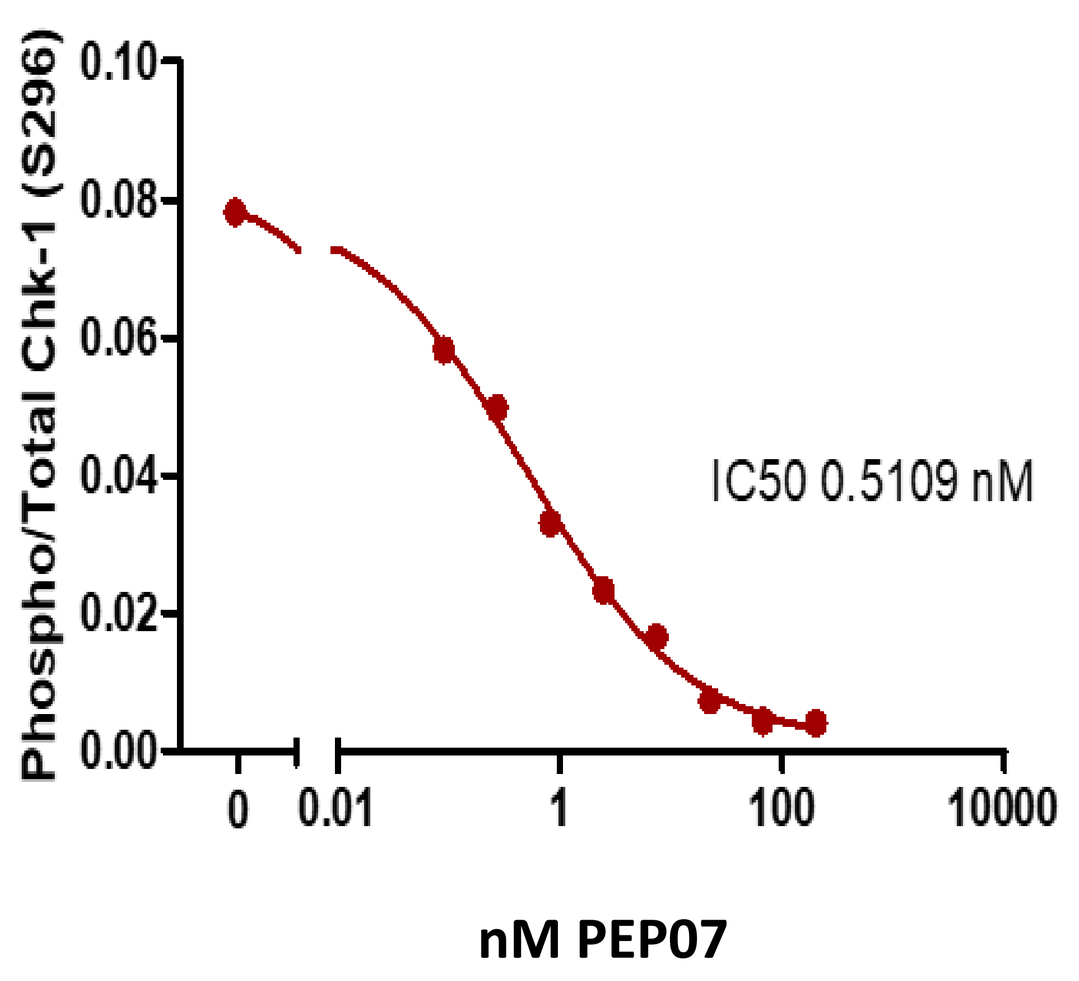
Phospho/Total Chk-1 (S296)
IC50 0.5109 nM
nM PEP07

C) d) e)

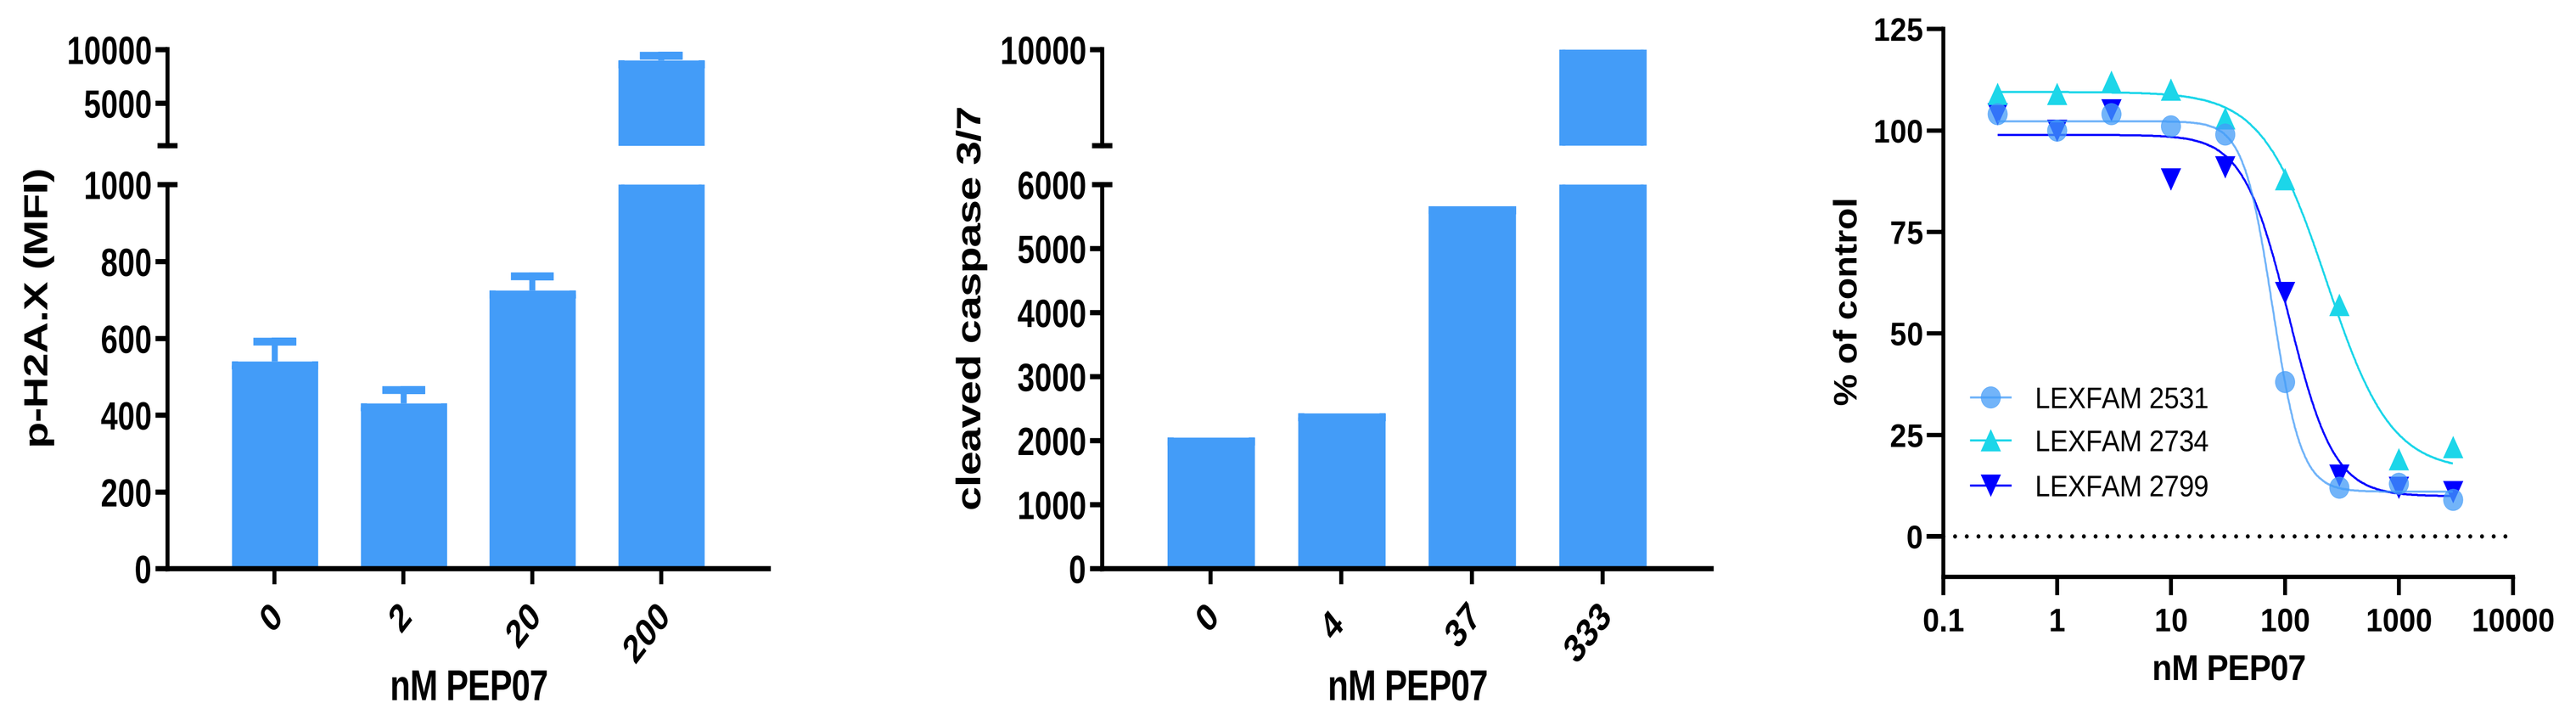
p-H2A.X (MFI)
nM PEP07
cleaved caspase 3/7
nM PEP07
% of control
LEXFAM 2531
LEXFAM 2734
LEXFAM 2799
nM PEP07

Figure 1. a) IC50s from ZLYTE FRET-based kinase assay b) Cellular IC50 as measured by autophosphorylation of pChk1 S296/total Chk1 in HT29 cells following 18h drug treatment c) phospho-H2A.X S139 in THP1 AML cell line following 18h drug treatment d) caspase 3/7 induction in Jeko-1 cell line following 18h drug treatment d) antiproliferative effects in AML patient derived cell models following 72h drug treatment.

PEP07 causes regressions in AML and MCL xenografts

PEP07 has been dosed orally as a single agent using a variety of dosing schedules, where it induces anti tumor activity and tumor regressions and is well tolerated.

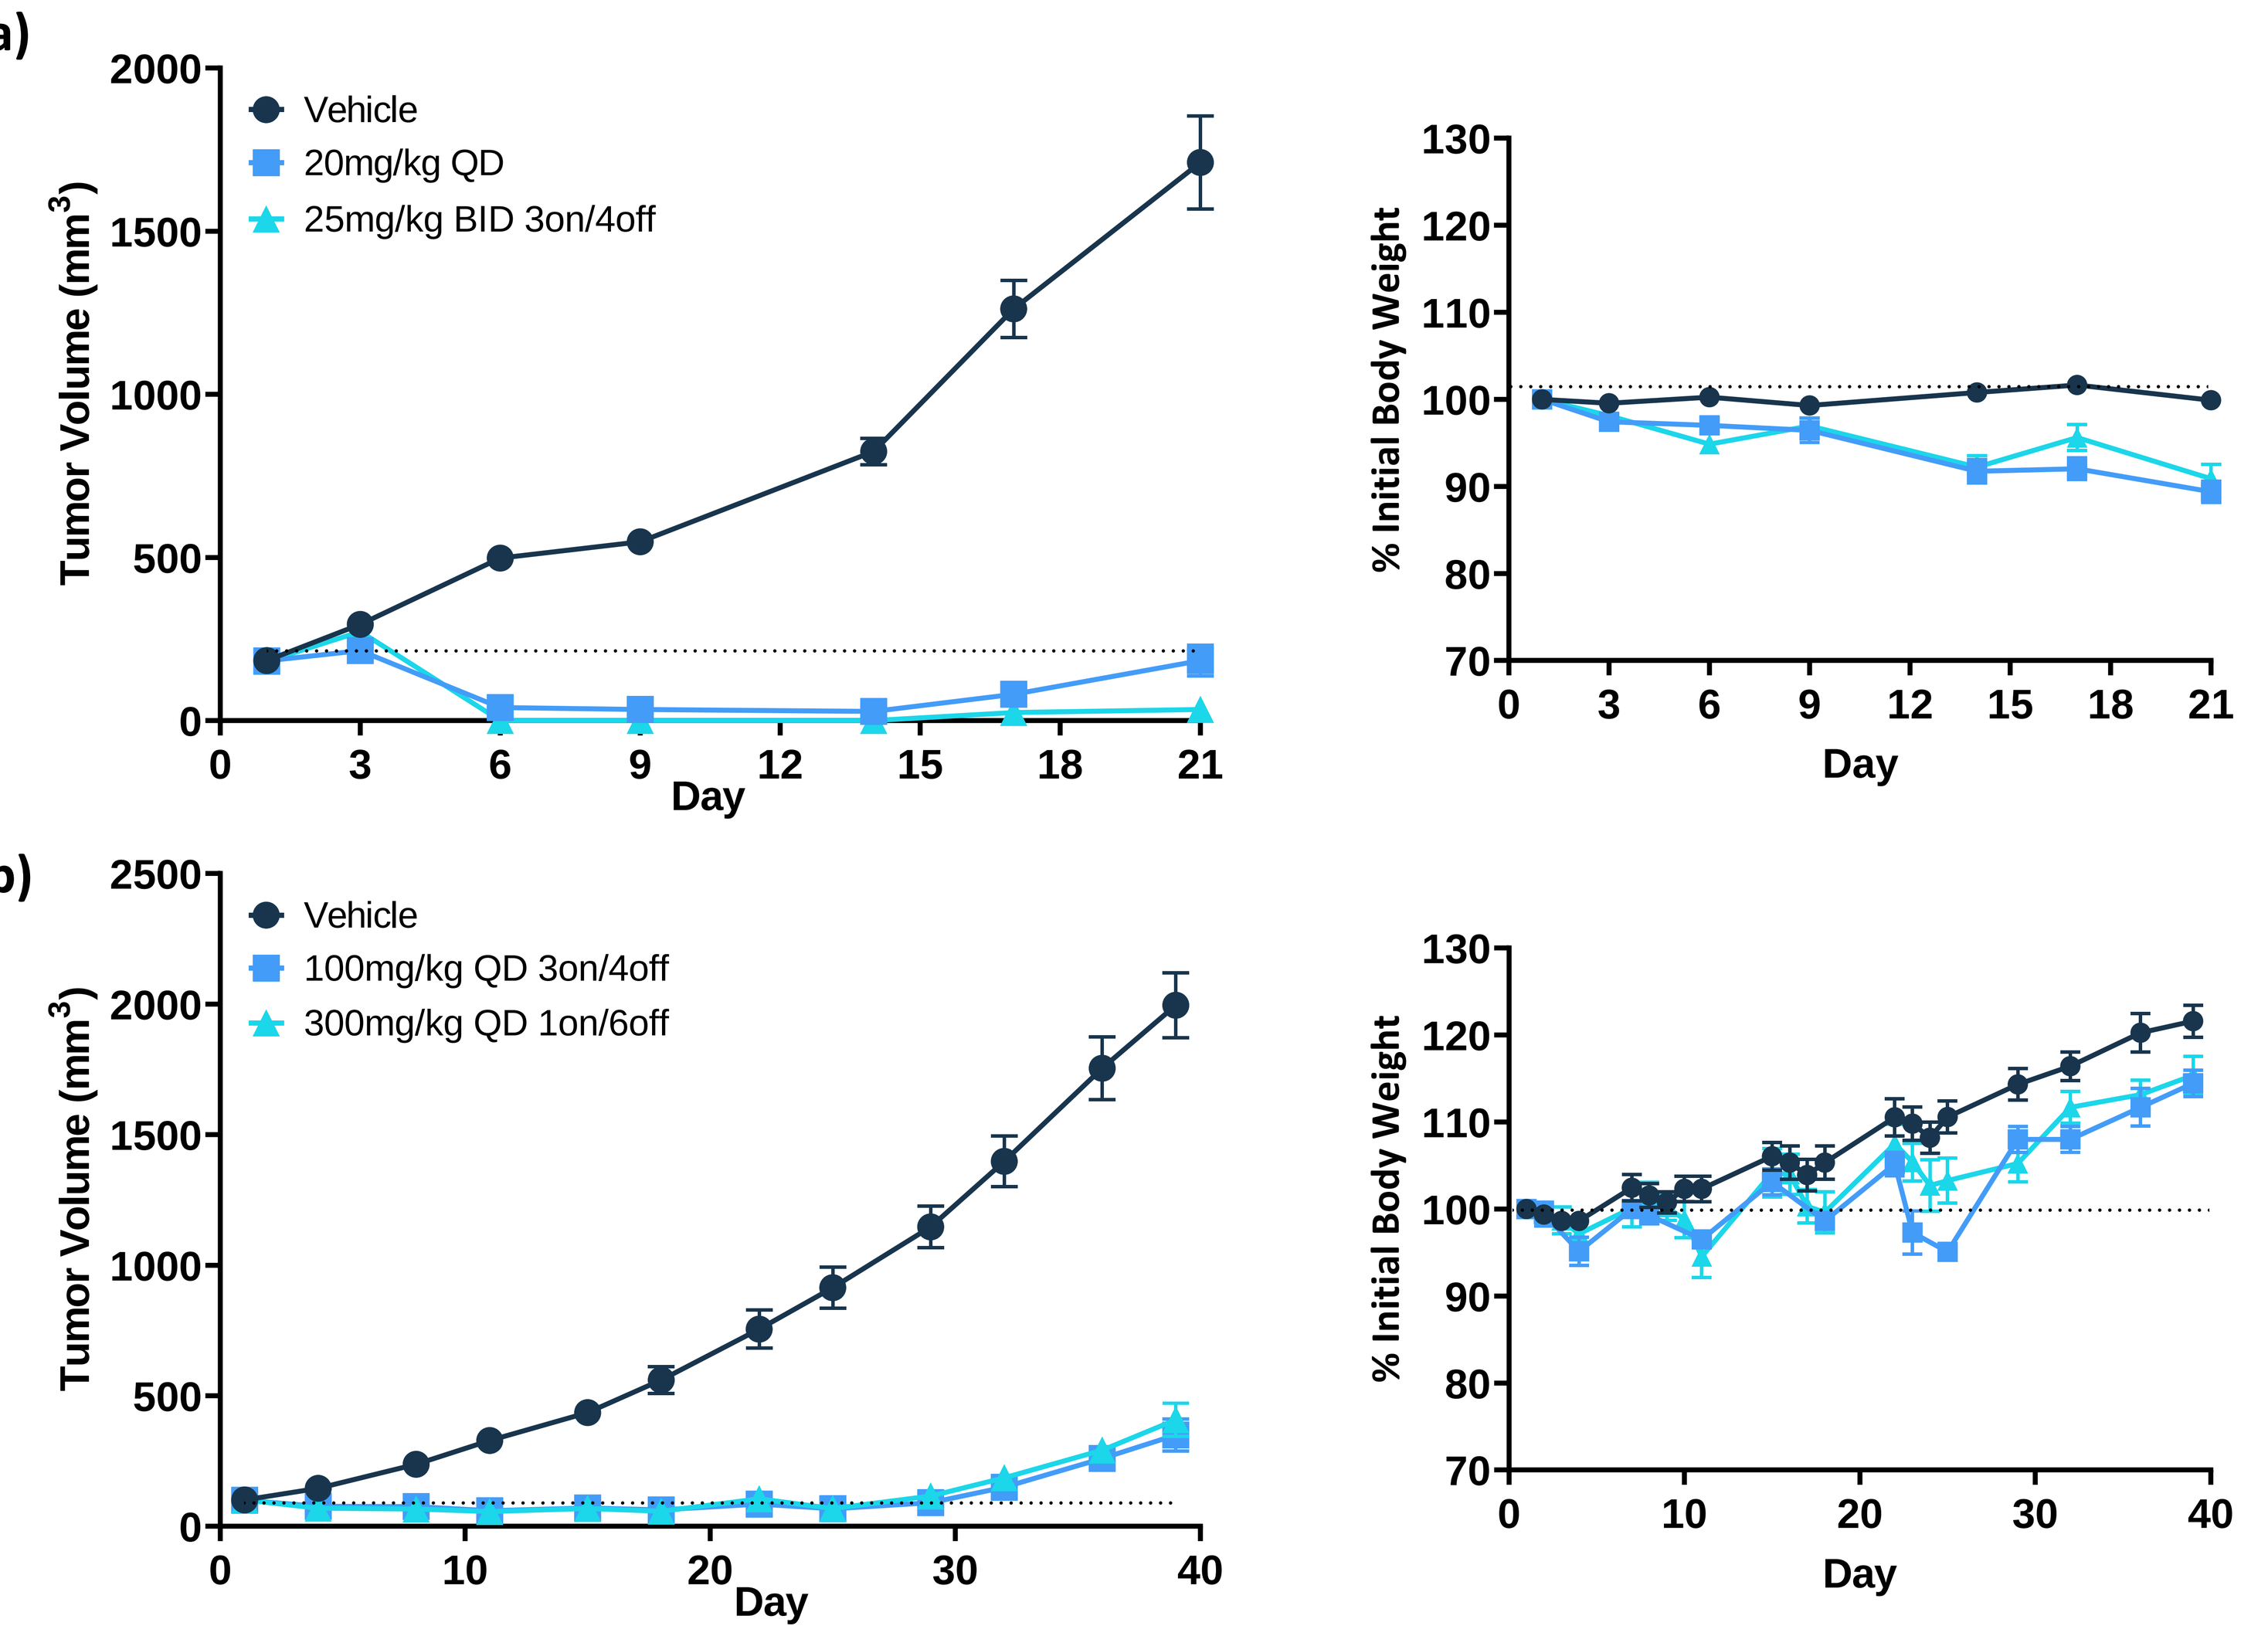
a)
Vehicle
20mg/kg QD
25mg/kg BID 3on/4off
Tumor Volume (mm³)
Day
% Initial Body Weight
Day
b)
Vehicle
100mg/kg QD 3on/4off
300mg/kg QD 1on/6off
Tumor Volume (mm³)
Day
% Initial Body Weight
Day

Figure 2. a) Jeko-1 MCL xenograft model. Mice were dosed with PEP07 daily for 21 days at 20mg/kg or BID 25mg/kg 3 days on followed by 4 days off. b) MV411 AML xenograft model. Mice were dosed with PEP07 once a week at 300mg/kg, or 100mg/kg for 3 days followed by 4 days off. Dosing was for 4 cycles.

PEP07 combines with cytarabine in AML xenografts

PEP07 combines with cytarabine in the cytarabine-sensitive MV411 model, and the cytarabine-insensitive THP1 model.

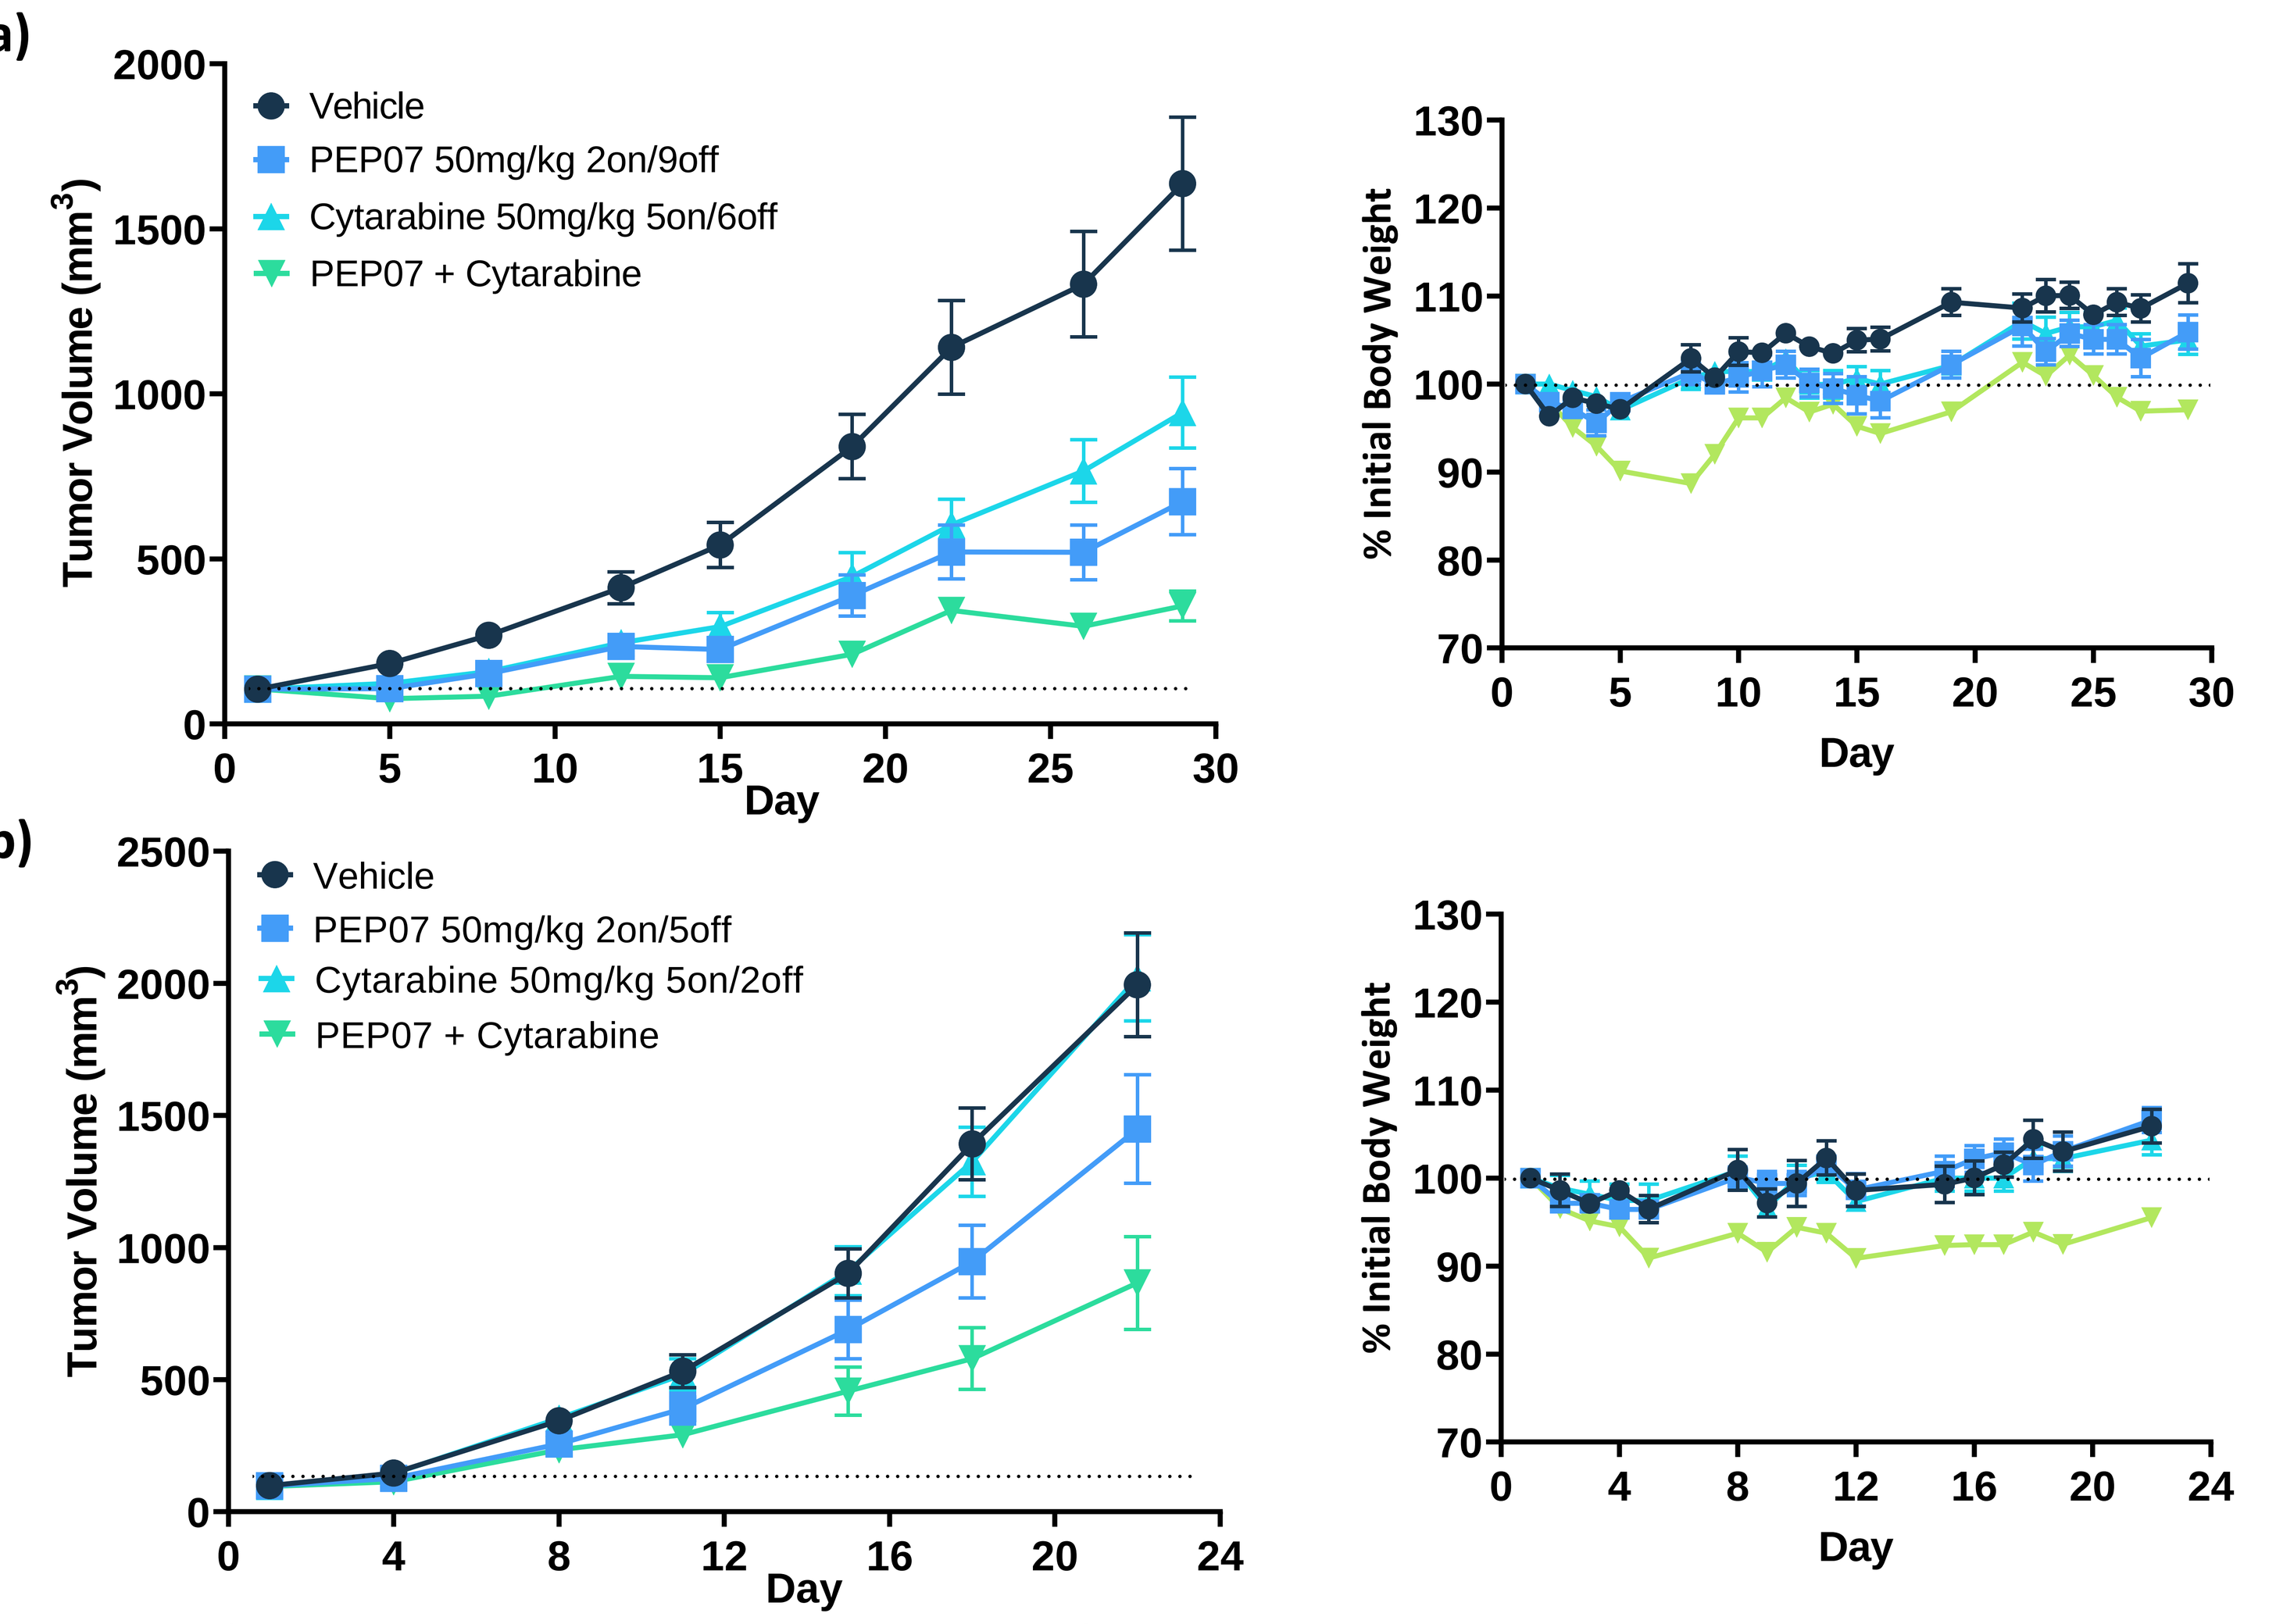
a)
Vehicle
PEP07 50mg/kg 2on/9off
Cytarabine 50mg/kg 5on/6off
PEP07 + Cytarabine
Tumor Volume (mm³)
Day
% Initial Body Weight
Day
b)
Vehicle
PEP07 50mg/kg 2on/5off
Cytarabine 50mg/kg 5on/2off
PEP07 + Cytarabine
Tumor Volume (mm³)
Day
% Initial Body Weight
Day

Figure 3. a) MV411 AML xenograft model and b) THP1 AML xenograft model, dosed as per the figure legends. PEP07 dosed PO and cytarabine dosed IP. Dosing was for 3 cycles.

PEP07 Tumor/Brain Penetration

Comparable exposures of PEP07 in the brain and plasma were observed in MV411 AML model, which suggests that PEP07 can penetrate the BBB in vivo. PEP07 significantly accumulates in the tumor.

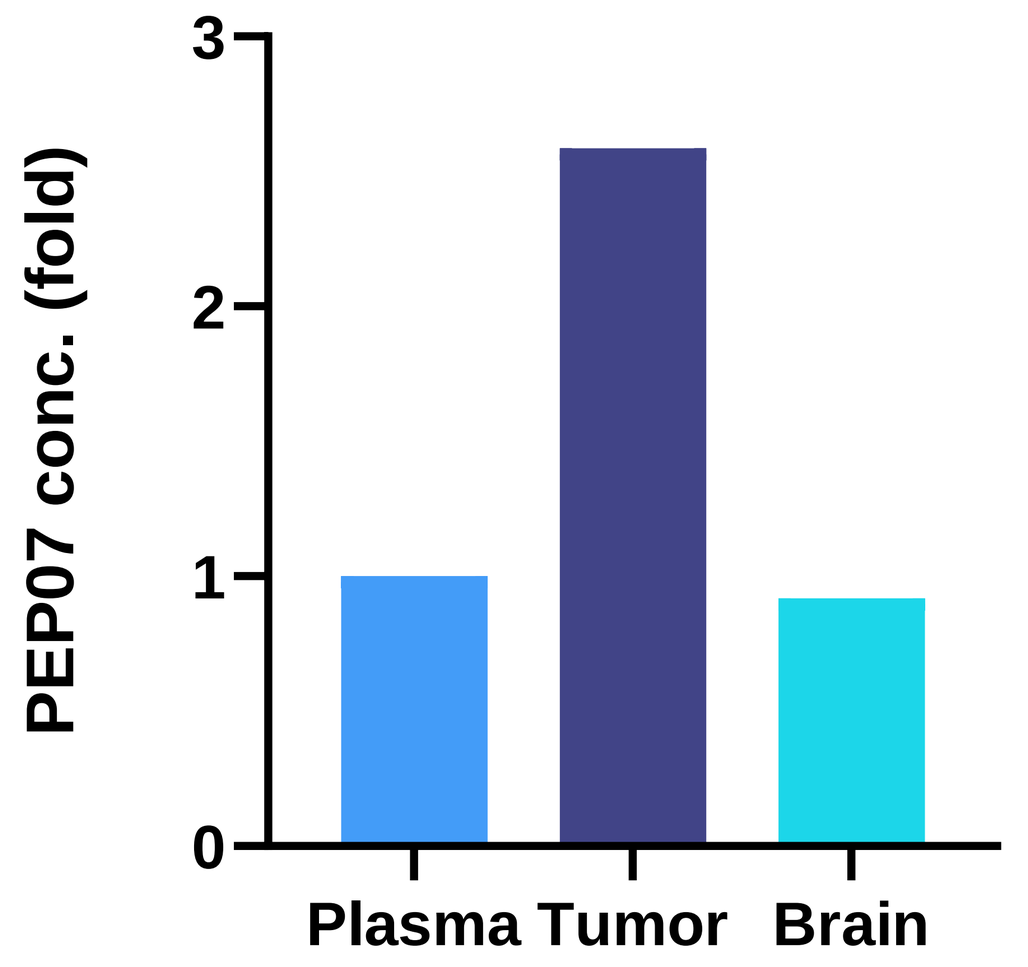
PEP07 conc. (fold)
Plasma
Tumor
Brain

Figure 4. Following a 6-week efficacy experiment, 4 mice were treated with PEP07 and samples collected 1h after dosing. LC-MS/MS analysis of PEP07 concentration in blood, tumor, and brain samples was conducted.

PEP07 Clinical Trial Design

The phase 1 clinical study for PEP07 comprises a dose escalation phase in relapsed/refractory (r/r) AML and MCL followed by a dose expansion phase in patients with r/r AML and/or selected tumor types. Patients will receive oral PEP07 treatment until disease progression or un-tolerable adverse event and DLTs will be evaluated during the first treatment cycle. Combination arms will follow to investigate PEP07 combination treatment in patients with r/r AML. The trial will be run in Australia and Taiwan which is aiming to begin patient recruitment in Q1 2023.

a) Study Design

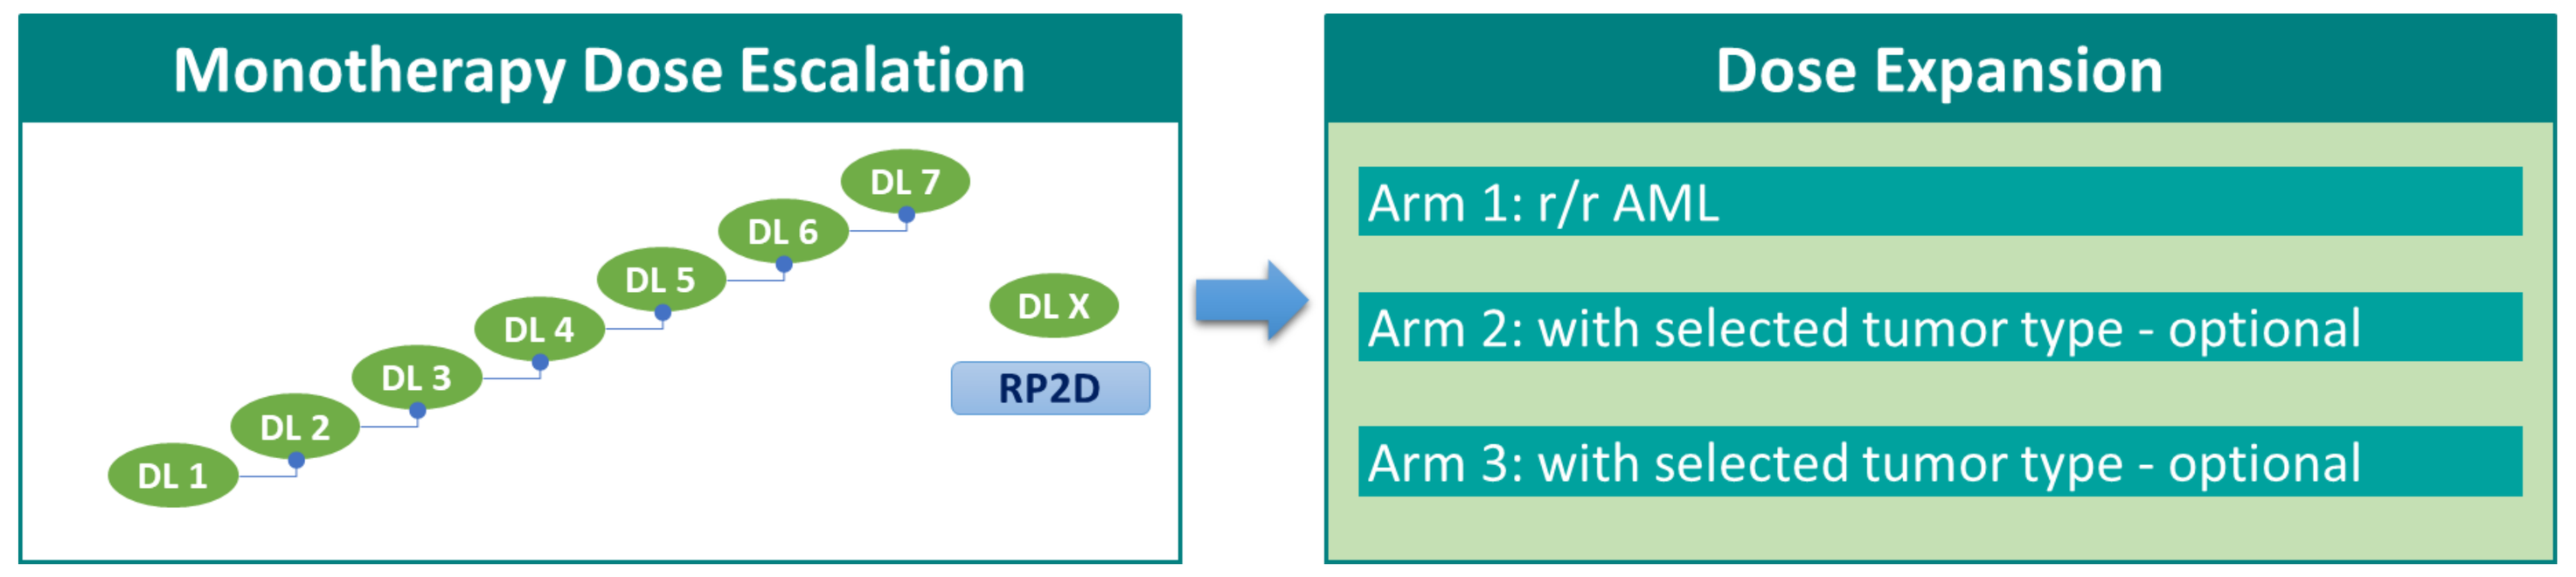
Monotherapy Dose Escalation
DL 1
DL 2
DL 3
DL 4
DL 5
DL 6
DL 7
DL X
RP2D
Dose Expansion
Arm 1: r/r AML
Arm 2: with selected tumor type - optional
Arm 3: with selected tumor type - optional

b) Study Schedule

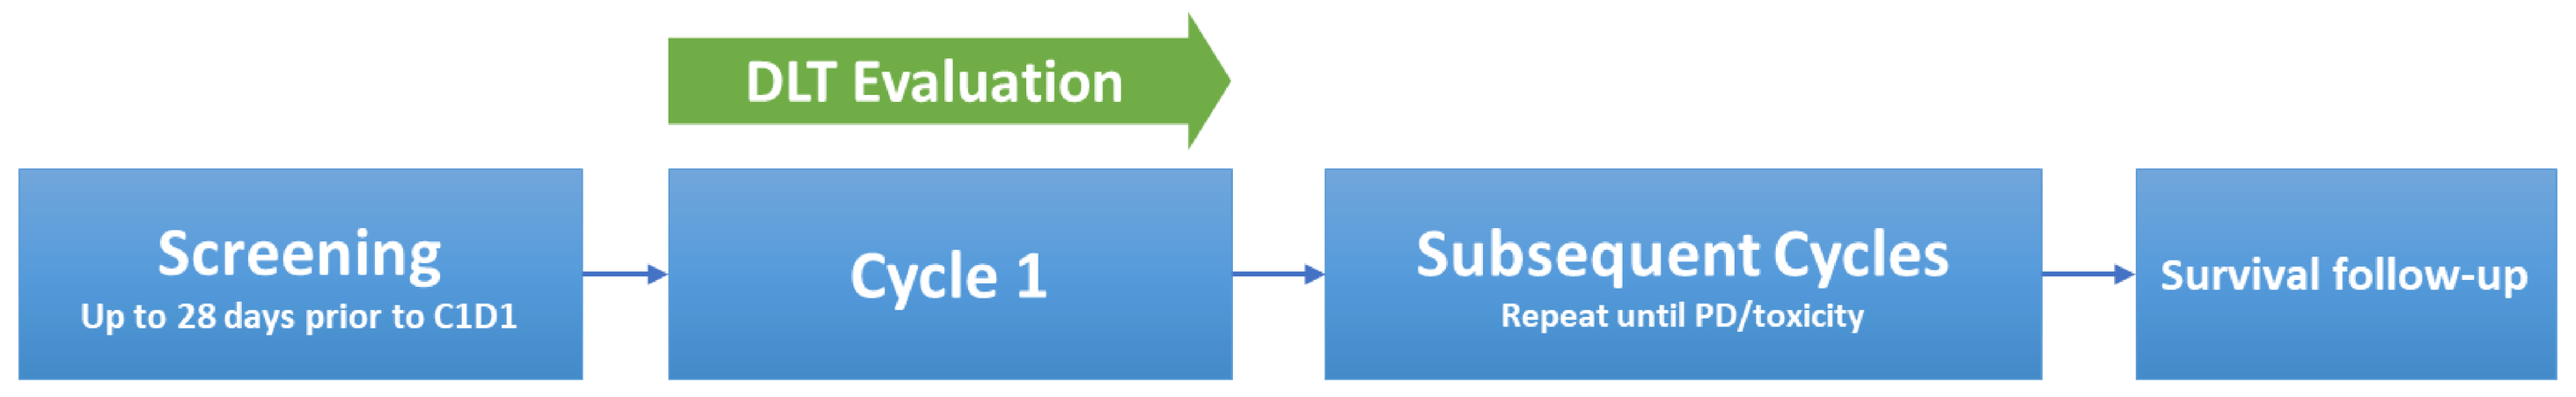
DLT Evaluation
Screening
Up to 28 days prior to C1D1
Cycle 1
Subsequent Cycles
Repeat until PD/toxicity
Survival follow-up

Figure 5. A schematic of the study design and schedule. a) Dose escalation and expansion with r/r AML and selected tumor, b) Study schedule contains screening, treatment and survival follow up period. The DLT is evaluated at cycle 1.

Conclusions

- PEP07 is a highly selective and potent Chk1 inhibitor that inhibits Chk1 autophosphorylation and induce apoptosis in cancer cell models
- PEP07 induces significant anti tumor efficacy as a single agent in models of AML and MCL
- PEP07 effectively combines with cytarabine in AML models
- PEP07 is advancing into Phase 1 clinical studies to treat patients with AML or MCL

Contact

Bettice Chen
PharmaEngine, Inc.
10 Minsheng E. Road, Sec.3 Taipei, Taiwan
bettice.chen@pharmaengine.com

Kyla Grimshaw
Sentinel Oncology Ltd
181, Cambridge Science Park, UK
kyla.grimshaw@sentineloncology.com

Author Affiliations

1. PharmaEngine, Inc. Taipei, Taiwan.
2. Sentinel Oncology Ltd, Cambridge, UK

References

3. Neizer-Ashun & Bhattachary, 2021. Cancer Lett, 497:202-211.
4. Chamoun & Borthakur, 2018. Expert Opin Investig Drugs, 27(8):661-666
5. Ferandez et al., 2005. J Clin Oncol, 23(26):6364-9

© Poster Template by Genigraphics® 1.800.790.4001 www.genigraphics.com